SUNDAY-434

La Trobe University, B.Hayes@latrobe.edu.au, +61 3 9479 3601

HXP124 as a novel treatment for onychomycosis

Brigitte Hayes[1], Mark Bleackley[1], Michael Baker[2], Pedro Quimbar[1], Marilyn Anderson[2], Nicole van der Weerden[2]
[1] La Trobe University, Melbourne, Australia, [2] Hexima Ltd., Melbourne, Australia

LA TROBE INSTITUTE FOR MOLECULAR SCIENCE

Introduction

Onychomycosis infections affect about 14% of the population[1], particularly the immunocompromised and the elderly[2]. The infections are most commonly caused by dermatophyte moulds, such as *Trichophyton rubrum* (45% of onychomycosis cases worldwide) and *Trichophyton interdigitale*[2,3].

Other non-dermatophyte moulds, such as *Scopulariopsis* and *Aspergillus spp.,* may also be present (10-15% of cases). Lastly, the yeast species *Candida albicans* and *Candida parapsillosis* may be the causative agents in patients with underlying predispositions[3]. Infections range from superficial to severe (Fig. 1).

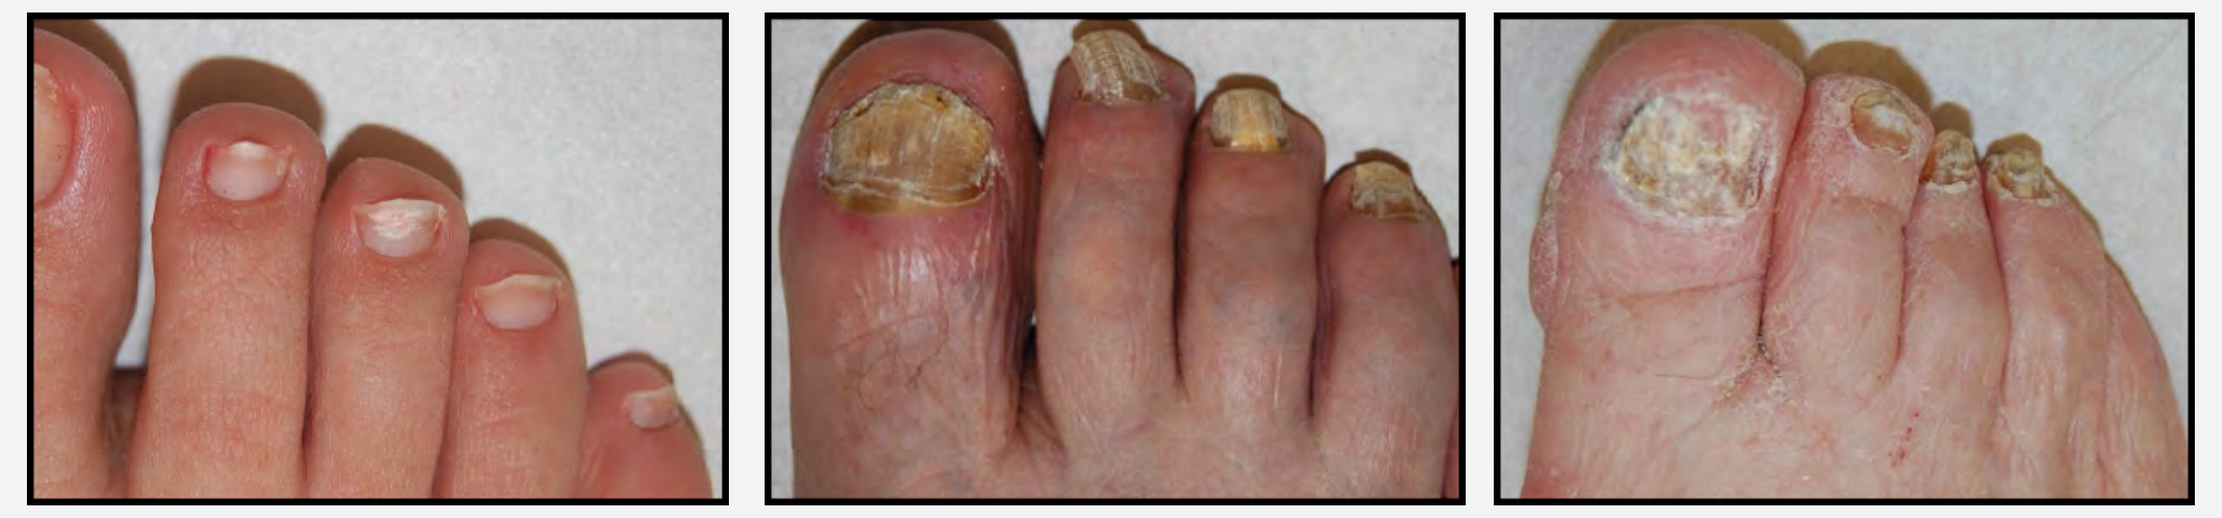

Figure 1. Onychomycosis infections range in severity
Toenails with onychomycosis infections of increasing severity (left to right) (Images from Ref. 2).

Onychomycosis infections can be treated with systemic therapies, such as the use of terbinafine or itraconazole. These antifungals are effective, but their usage is limited by drug-drug interactions and adverse effects. Systemic treatment with terbinafine may result in liver toxicity, while treatment with itraconazole poses the risk of ventricular dysfunction[3]. Topical treatments have a lower risk for adverse effects but are complicated by the need for the drug to penetrate the nail (Fig. 2).

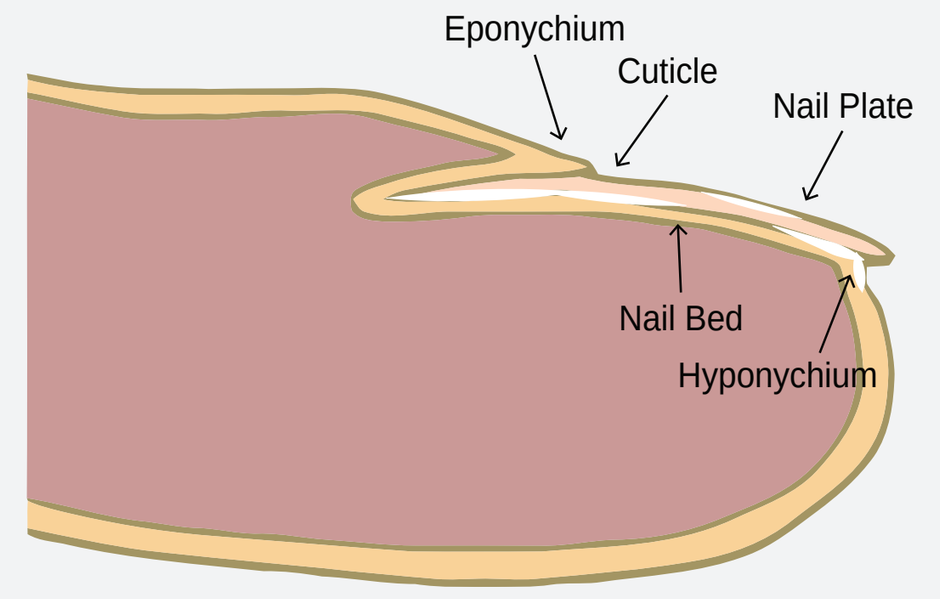
Eponychium
Cuticle
Nail Plate
Nail Bed
Hyponychium

Figure 2. Nail cross section
A cross section of a nail showing common areas of fungal growth (white areas)[4].

Current topical therapies have poor penetration rates (Fig. 3)[5,6], resulting in lower cure rates than for systemic treatments (Table 1)[3]. Ciclopirox has extremely low penetration rates (e.g 0.03% penetration on Day 3 of dosing) and has cure rates from 6 - 9%. Tavaborole has the highest penetration of the current topical drugs with 15% cumulative penetration by Day 15. However cure rates are only slightly higher at 7 - 9%. The most recent of these drugs, efinaconazole, has a slightly higher cure rate at 15-18%. In addition, the current therapies require long treatment times (48 weeks) and may also depend on physical debridement of the nail[3,7].

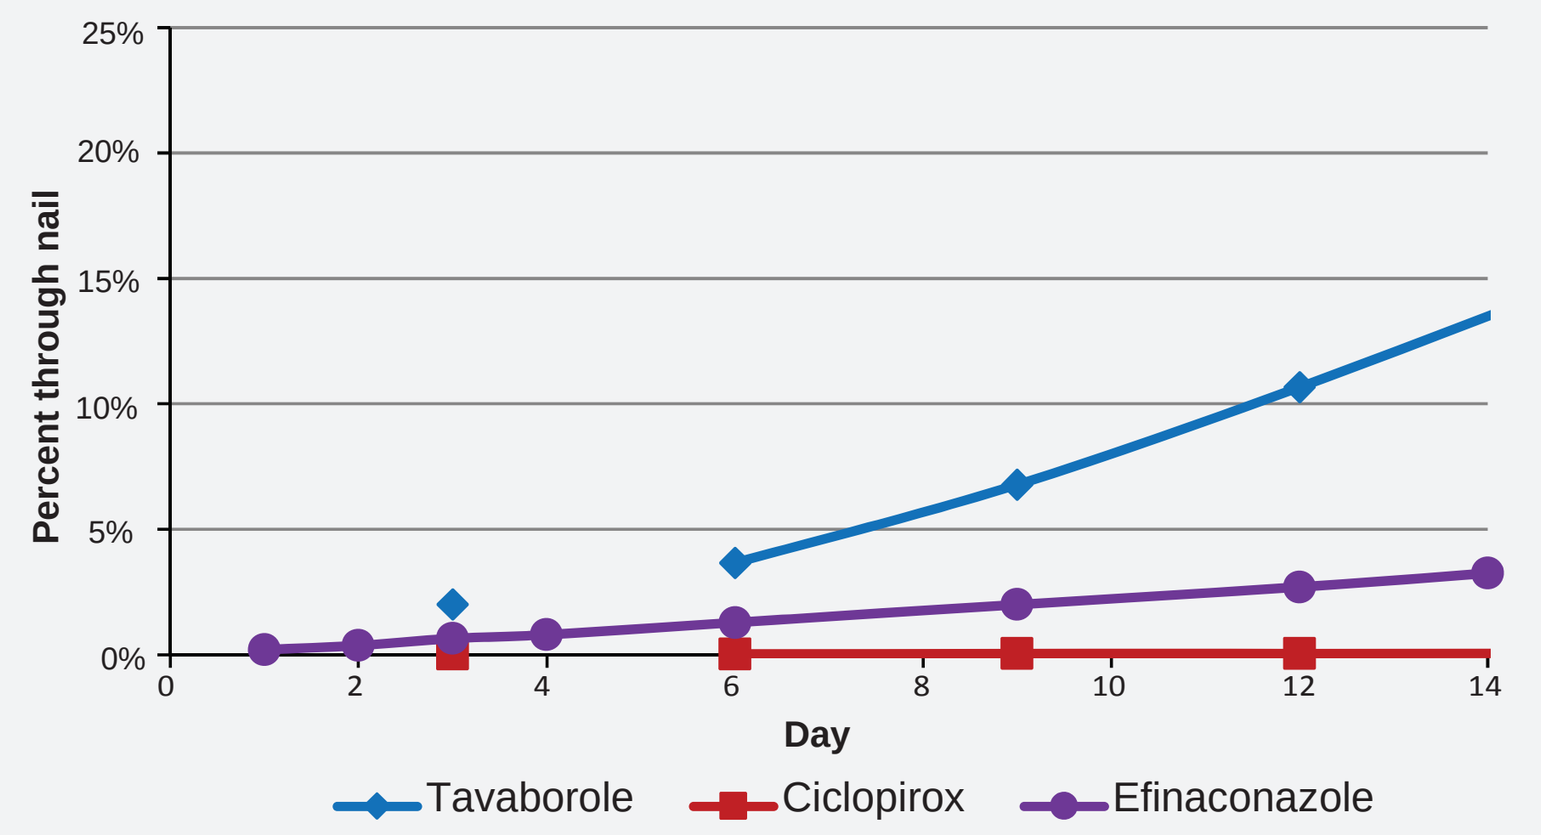
Percent through nail
Day
Tavaborole
Ciclopirox
Efinaconazole

Figure 3. Cumulative percent penetration of nails by current topical therapies
Cumulative penetration of nails by ciclopirox (red), tavaborole (blue) and efinaconazole (purple)[5,6].

Table 1. Complete cure rates of current topical therapies[3]

Treatment	Complete cure rate
Ciclopirox (Penlac™)	6 - 9%
Tavaborole (Kerydin™)	7 - 9%
Efinaconazole (Jublia©)	15-18%

Methods & Results

The plant defensin HXP124 is a potential onychomycosis treatment

In comparison to current topical therapies, which are all small hydrophobic molecules, we have focused on the use of an antuifungal peptide for the treatment of onychomycosis. Most living organisms express antimicrobial peptides as part of their innate immune sytem. The plant defensins are of particular interest due to their potent antifungal activity. Plant defensins are small (~ 5 kDa), basic proteins with four disulfide bonds. Despite great variation in amino acid sequence they share a common tertiary structure, consisting of a triple-stranded, anti-parallel β-sheet and an α-helix (Fig. 4)[8].

The hydrophillic nature of plant defensins may be more suited to traversal of the nail plate, allowing access to the fungus growing underneath (Fig. 1B). A plant defensin, HXP124, was selected as a potential onychomycosis topical treatment. This peptide was selected for its potent antifungal activity against the fungal species responsible for onychomycosis and its ability to efficiently cross the nail plate.

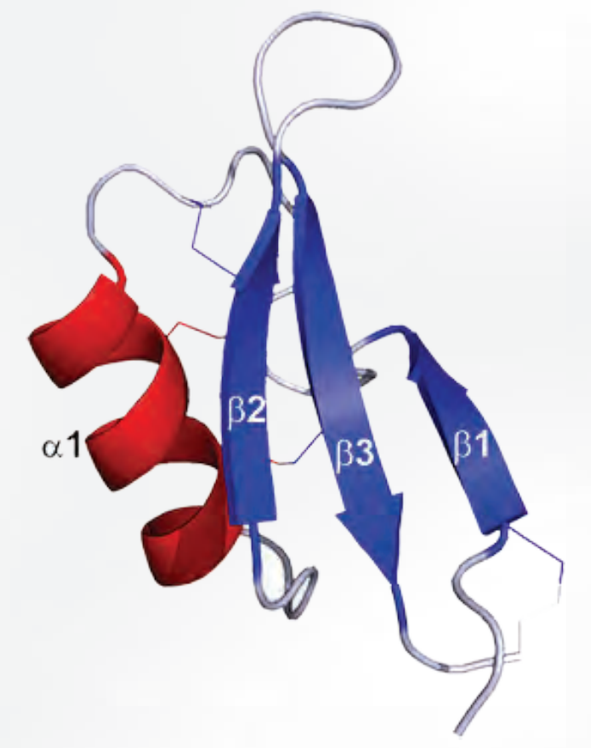
α1
β2
β3
β1

Figure 4. Tertiary structure of the plant defensin NaD1 (PDB: 1MR4).

HXP124 is a potent antifungal molecule against dermatophyte moulds

HXP124 has fungal inhibitory activity (at low μg/mL concentrations) against the pathogens most commonly associated with onychomycosis. HXP124 was most active against *T. rubrum*, the species which generally causes these infections (Table 2). The inhibitory activity of HXP124 against *T. rubrum* was comparable to the activity of drugs currently used as topical treatments for onychomycosis (Table 3).

Table 2. Growth inhibiton (IC_{50}) of fungal species associated with onychomycosis by HXP124
Fungal spores were incubated overnight with various concentrations of HXP124 (0 - 100 μg/mL). IC_{50} values were obtained by determining the HXP124 concentration at which 50% inhibition was achieved. Data is from at least three independent experiments.

Fungal Species	IC_{50} (μg/mL) ± SD
Trichophyton rubrum	**3.7** ± 1.5
Trichophyton interdigitale	**5.8** ± 2.0
Microsporum fulvum	**9.6** ± 2.5
Aspergillus niger	**35.2** ± 9.5
Candida albicans	**10.3** ± 3.0

Table 3. Fungal inhibiton (IC_{50}) of *T. rubrum* by HXP124 compared to other topical therapies.
Fungal spores were incubated overnight with various concentrations of HXP124 (0 - 100 μg/mL). IC_{50} values were obtained by determining the concentration at which 50% inhibition was achieved. Data is from at least two independent experiments.

Treatment	IC_{50} (μg/mL) ± SD
HXP124	**3.7** ± 1.5
Ciclopirox olamine	**0.5** ± 0.11
Tavaborole	**5.3** ± 0.40

HXP124 rapidly kills fungal cells

HXP124 kills 20% and 90% of *C. albicans cells* when treated for only 30 min with 10 and 50 μg/mL HXP124 respectively (Fig. 5). For *T. rubrum*, survival was reduced to 2% after 24H incubation with 50 μg/mL HXP124. *T. rubrum* survival continues to drop with additional 50 μg/mL doses (Fig. 6A). *T. rubrum* survival was also reduced with 10 μg/mL HXP124 treatment. However, 24 days of daily treatment was required to reach 5% survival. After 16 days of daily dosing of a *T. rubrum* liquid culture, the HXP124 treated cultures were clear to the eye, while the untreated control had dense growth (Fig. 6B).

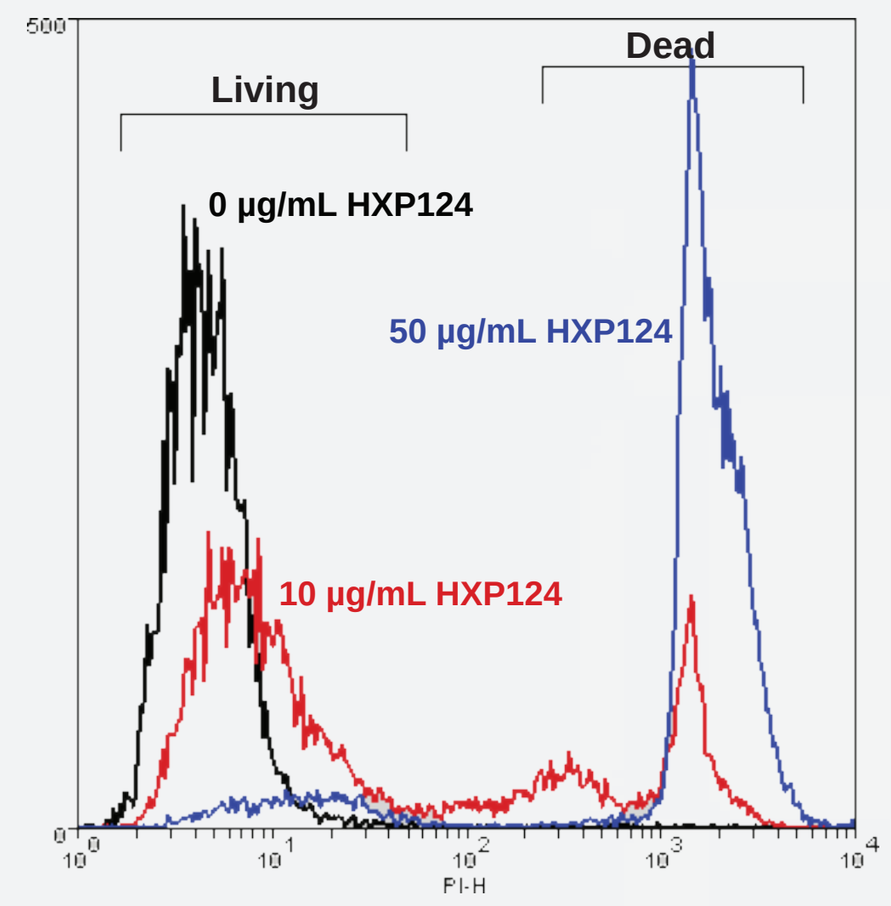
Living
Dead
0 μg/mL HXP124
50 μg/mL HXP124
10 μg/mL HXP124
PI-H

Figure 5. HXP124 kills *C. albicans*
C. albicans cells were treated for 30 min with 0, 10 and 50 μg/mL HXP124. The death marker propidium iodide (PI) was added to the cells and the number of PI positive cells were counted using flow cytometry.

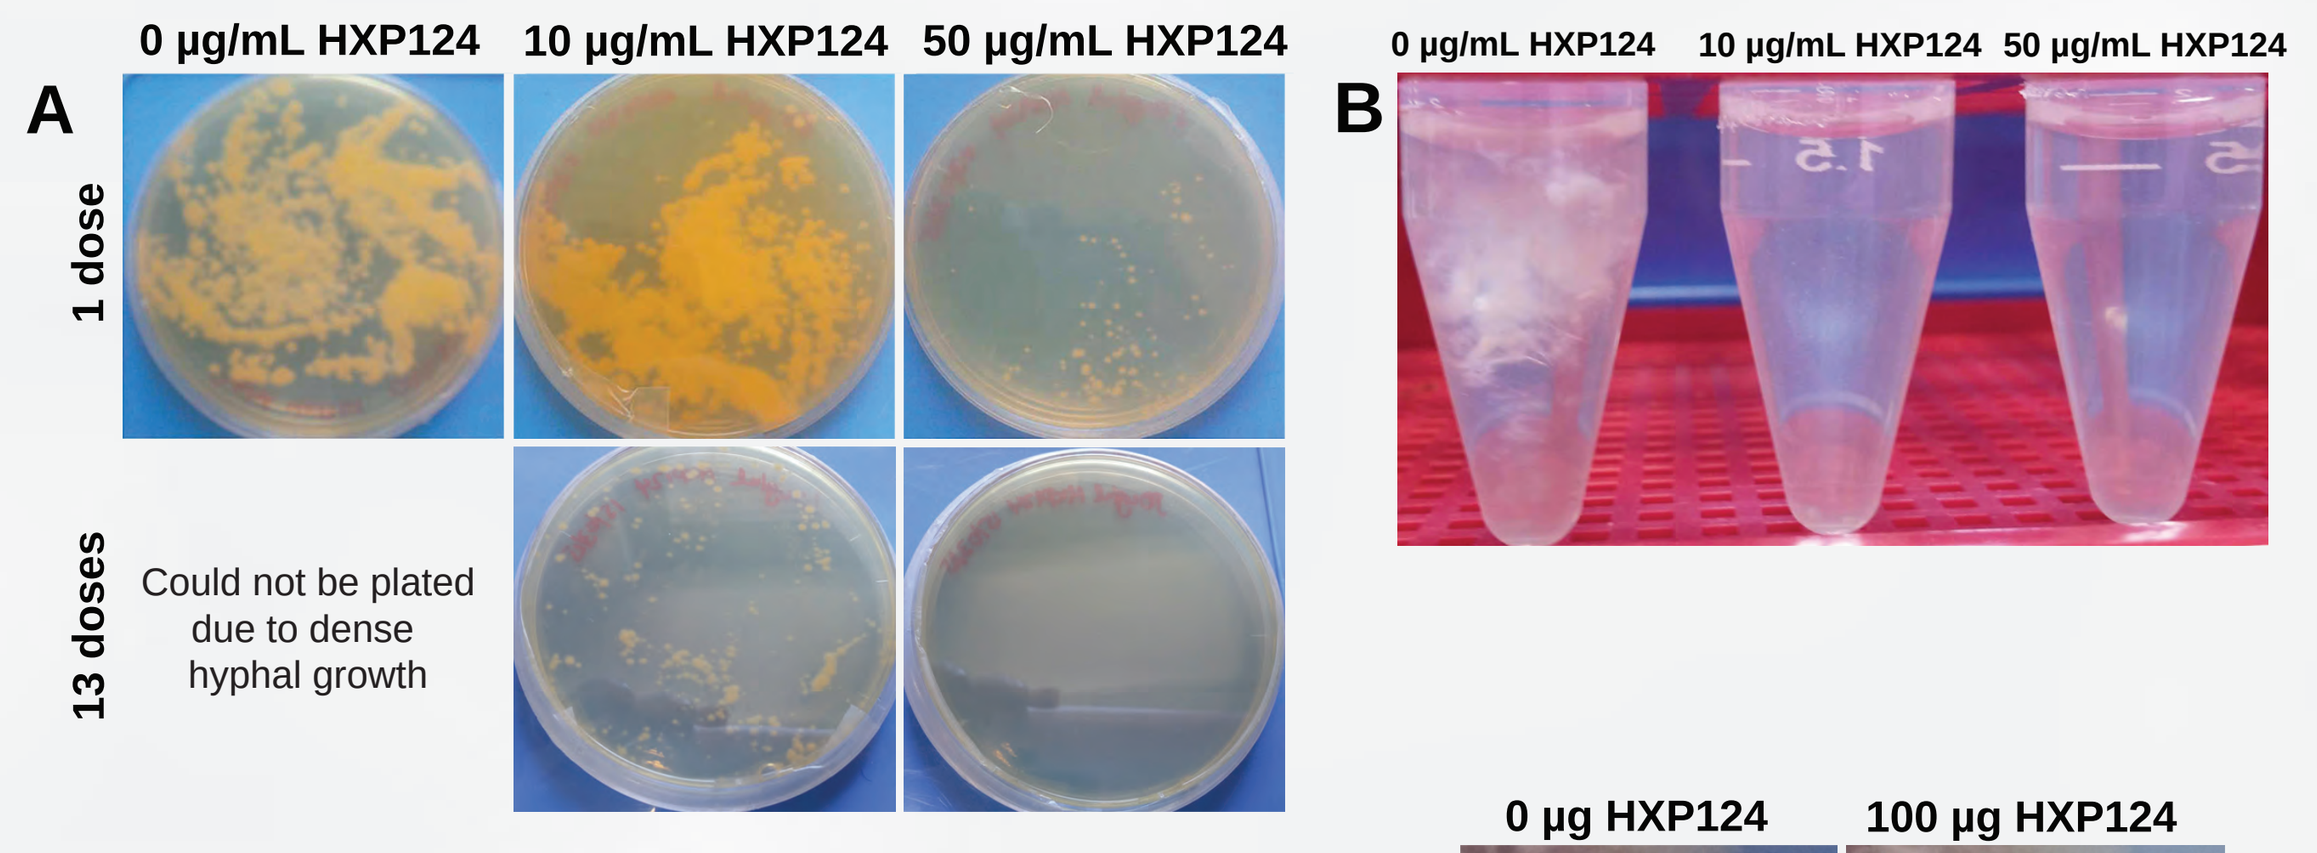
A
0 μg/mL HXP124
10 μg/mL HXP124
50 μg/mL HXP124
1 dose
13 doses
Could not be plated due to dense hyphal growth
B
0 μg/mL HXP124
10 μg/mL HXP124
50 μg/mL HXP124

Figure 6. Survival of *T. rubrum* treated daily with HXP124
T. rubrum spores (in 2 mL of ½ PDB culture) were treated daily with 0, 10 or 50 μg/mL HXP124. The following day an aliquot was plated onto ½ SDA and and grown to check for surviving colonies (CFU).
(A) *T. rubrum* CFU at 24 H and 13 days of treatment with 0, 10 or 50 μg/mL HXP124.
(B) Liquid cultures after 16 days of treatment with 0, 10 or 50 μg/mL HXP124.

HXP124 also retains activity when *T. rubrum* is grown on keratin

Many dermatophytes produce keratinases that allow them to grow on keratin as the sole carbon and nitrogen source[9]. Clear zones of inhibition were observed with multiple HXP124 concentrations, when *T. rubrum* was grown on ½ PDA (a nutrient rich agar on which *T. rubrum* would not require keratinases). HXP124 also inhibited growth when 1% keratin was used as the sole nutrient source (Fig. 7).

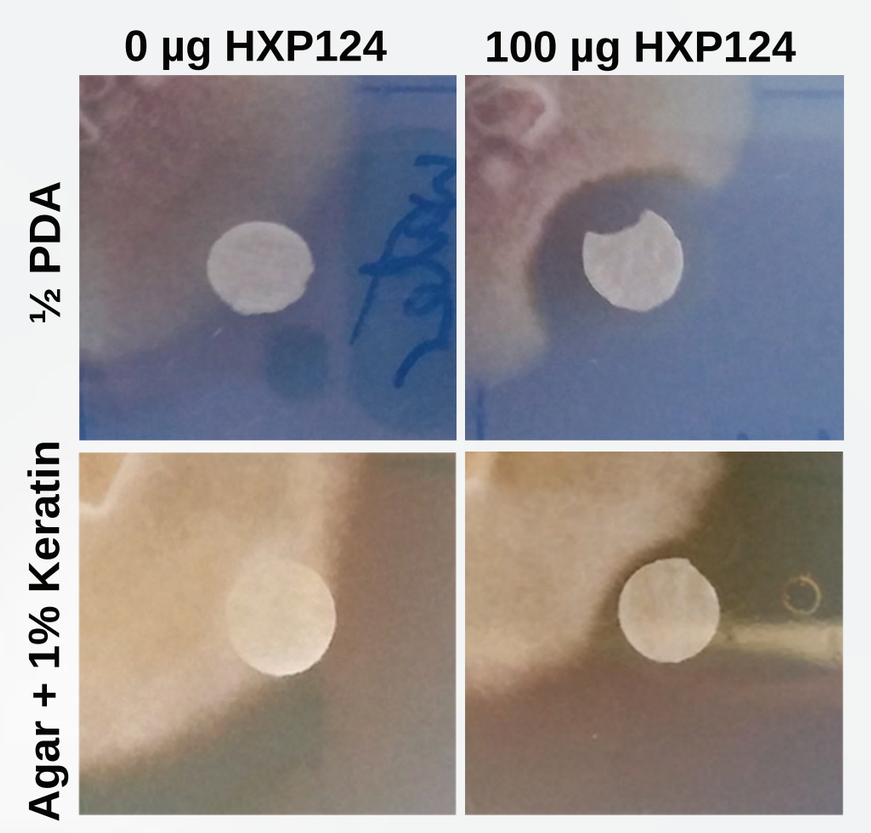
0 μg HXP124
100 μg HXP124
½ PDA
Agar + 1% Keratin

Figure 7. HXP124 activity against *T. rubrum* grown on keratin
Filter disks were soaked in HXP124 and placed on various agar types. *T. rubrum* was placed in the center of the plate and incubated until the growth had reached the filter disks. Zones of inhibiton of *T. rubrum* growth were observed after exposure to 100 μg HXP124 on ½ PDA or Agar with 1% Keratin as the sole nutrient source.

HXP124 does not bind to keratin

HXP124 remained in the supernatant rather than the keratin pellet for all HXP124 concentrations tested, when incubated with insoluble keratin, indicating that HXP124 was unable to bind keratin (Fig. 8).

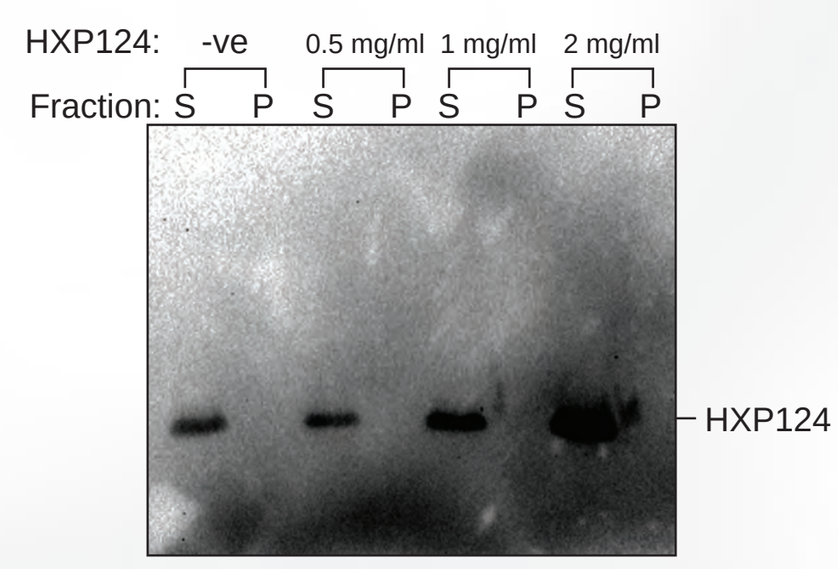
HXP124: -ve 0.5 mg/ml 1 mg/ml 2 mg/ml
Fraction: S P S P S P S P
HXP124

Figure 8. HXP124 does not bind keratin
Multiple HXP124 cocentrations were incubated with insoluble keratin (50% in water). The mixutre was then spun down and a Western blot performed with α-HXP124 antibodies to determine if HXP124 was retained in the keratin pellet.

HXP124 is able to efficiently penetrate the nail plate

Large amounts of HXP124 penetrated human fingernail clippings when 10 μL of a 1% HXP124 solution was added daily to nails (Fig. 9). An average of 4% HXP124 penetrated the nails on the first day and penetration continued to increase over subsequent days (Fig. 9A). After 14 days of dosing, a cumulative average of 23% had passed through the nails (Fig. 9B).

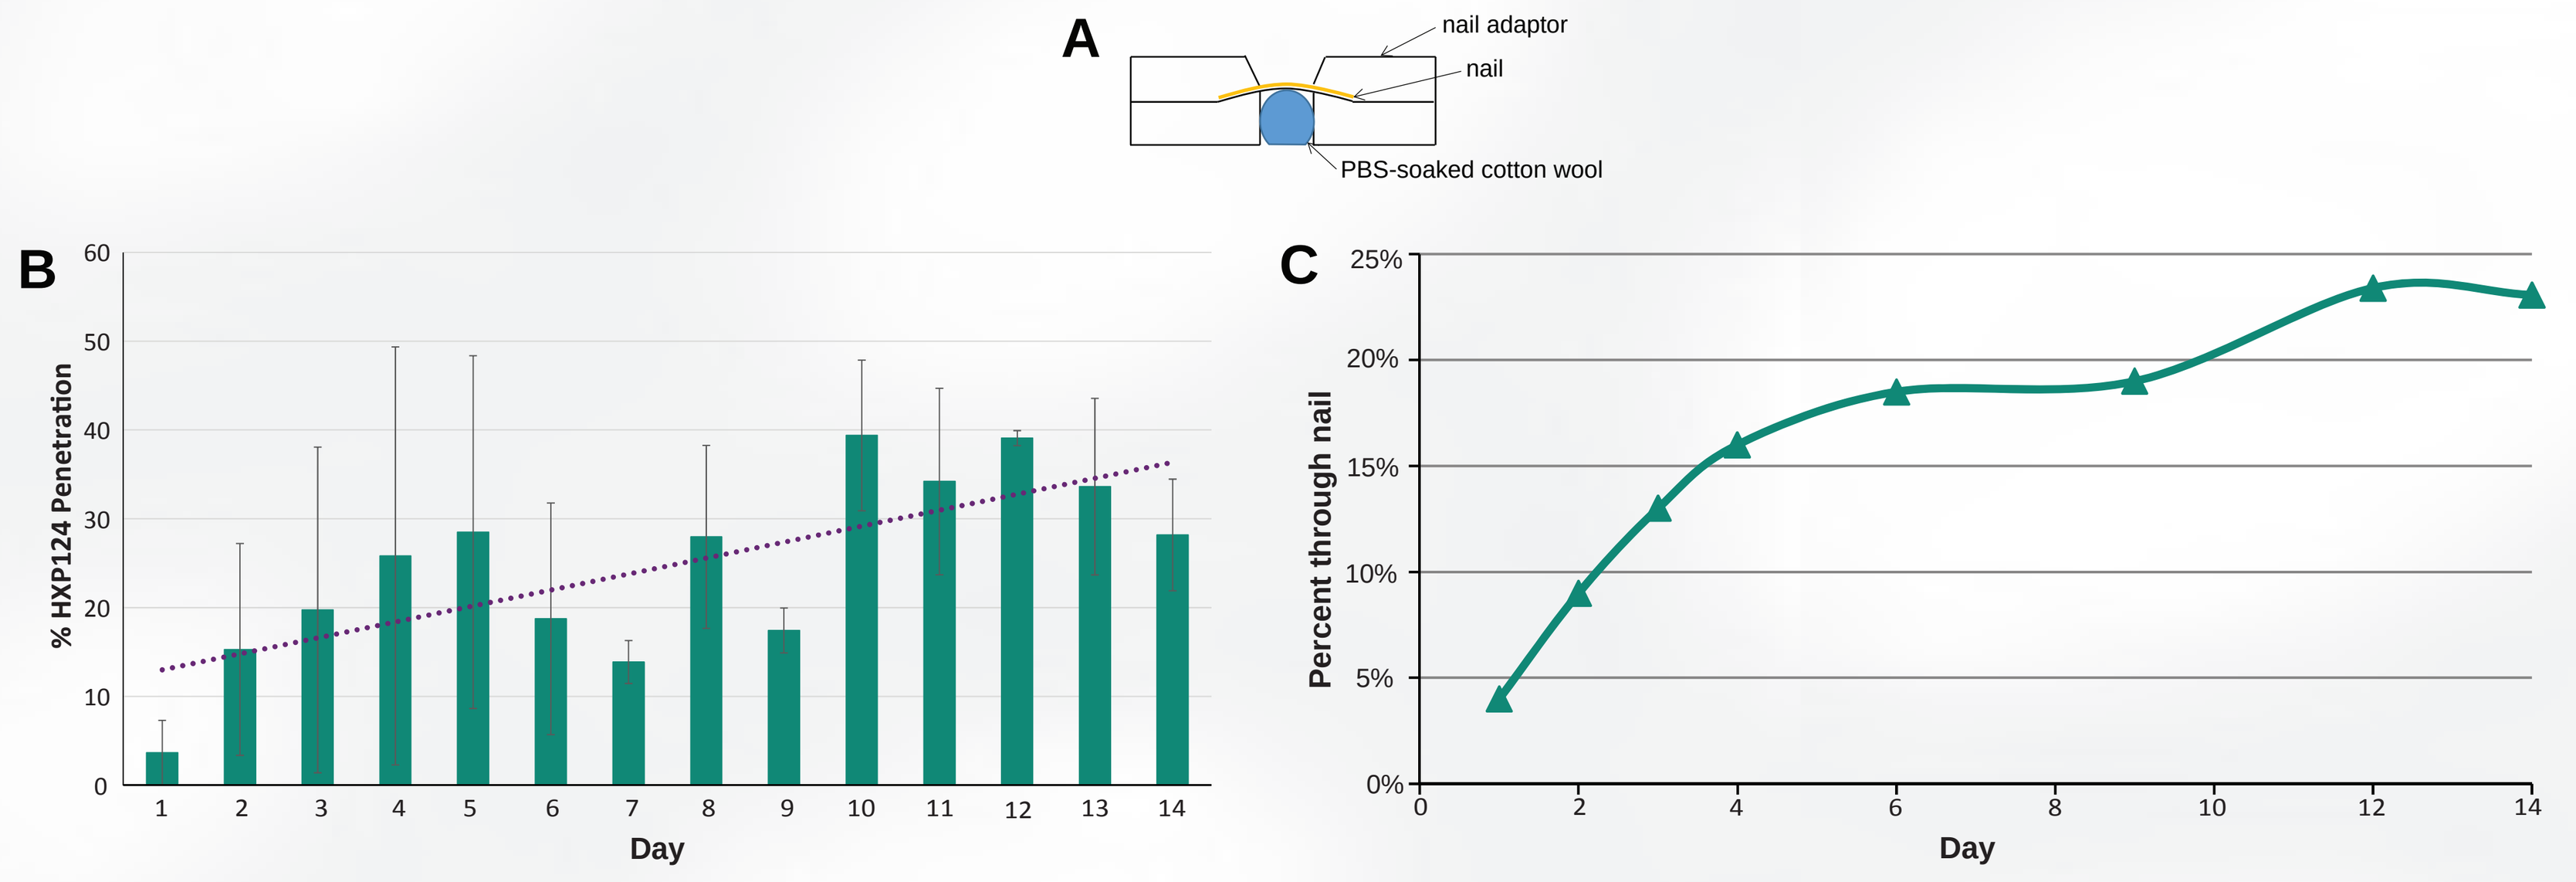
A
nail adaptor
nail
PBS-soaked cotton wool
B
% HXP124 Penetration
Day
C
Percent through nail
Day

Figure 9. HXP124 penetration of human nails
Human nails were placed into specialised nail adaptors (**A**, PermaGear) and PBS-soaked cotton wool was placed in contact with the nail underneath. Each day, a single 10 μL dose of a 1% HXP124 solution was added to the top of the nail and incubated overnight at 26°C. The following day the top of the nail was washed and the cotton wool replaced. Each day the contents of the cotton wool were removed by washing and then HXP124 in the resulting wash was quantified by RP-HPLC.
(B) Average percentage HXP124 penetration through fingernail clippings daily for 14 days of dosing with 1% HXP124. Error bars are SEM (n=3).
(C) Cumulative percentage HXP124 penetration through fingernail clippings over 14 days of dosing with 1% HXP124 (n=3).

HXP124 retains activity after passing through the nail

Penetration of nails does not effect the growth inhibitory activity of HXP124. A fungal inhibition assay against *C. albicans* with HXP124 collected under the nail in penetration assays had a similar level of activity to a fresh HXP124 stock (Fig. 10).

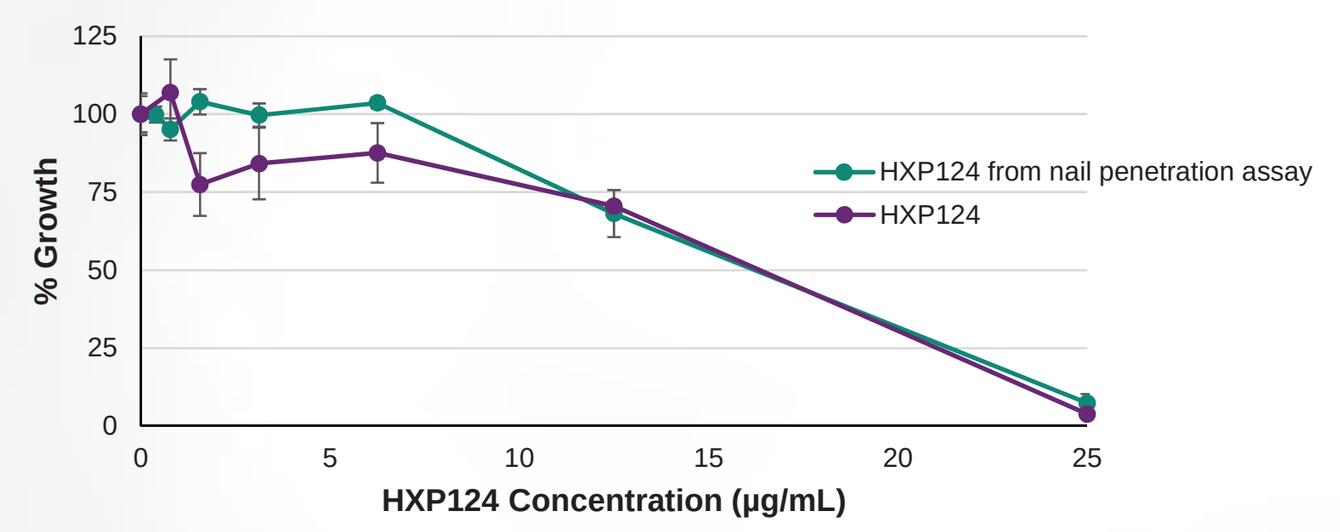
% Growth
HXP124 Concentration (μg/mL)
HXP124 from nail penetration assay
HXP124

Figure 10. HXP124 activity is not effected by nail penetration
A fungal inhibition assay with HXP124 collected from a penetration assay compared to HXP124 alone. Data was converted to percentage growth relative to a no-protein control. Error bars are SD from three technical replicates.

Conclusions

HXP124 has inhibitory activity against the fungal species that cause nail infections and retains this activity in the presence of keratinases. Importantly, it has similar levels of activity to the current therapeutics. HXP124 is also fungicidal, not fungistatic, meaning it kills rather than inhibiting growth. This could potentially be an advantage compared to fungistatic families of drugs like the azoles (of which efinaconazole is a member), which require cell turnover to clear the fungus and have an increased risk of resistance developing[10]. HXP124 also has increased nail penetration in comparison to current topical therapies, with greater than 85% improvement compared to efinaconazole, the drug with the highest cure rate of the current therapies (Fig. 11).

Together, this data shows that HXP124 has the potential to be an improved topical therapeutic for the treatment of onychomycosis, with greater efficacy and shorter treatment times than the current topical therapeutics.

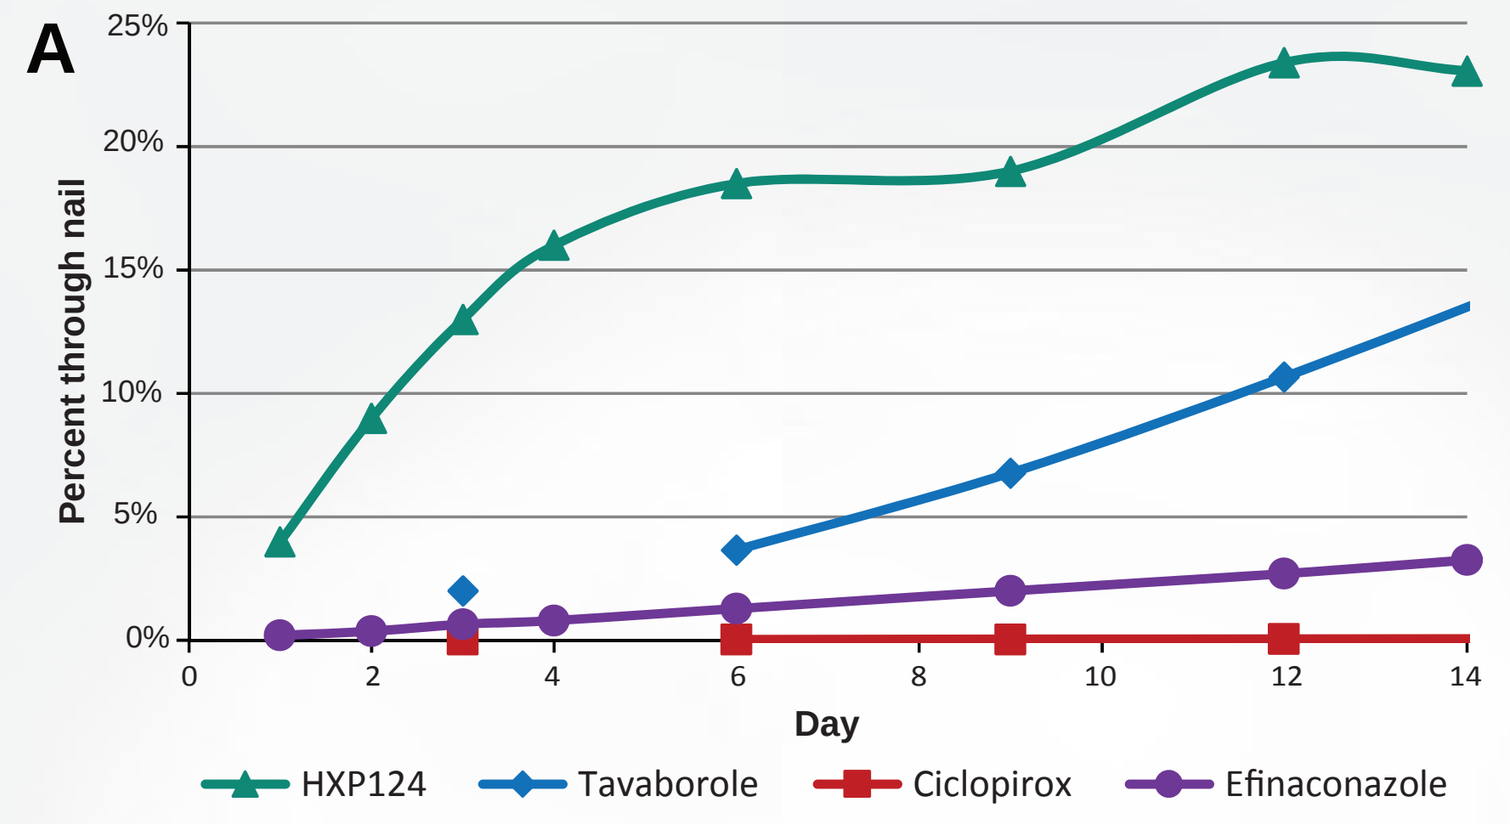
A
Percent through nail
Day
HXP124
Tavaborole
Ciclopirox
Efinaconazole

B

Treatment	Permeation relative to HXP124
HXP124	100%
Ciclopirox	0.21%
Tavaborole	45.58%
Efinaconazole	11.54%

Figure 11. Cumulative percent penetration of nails by HXP124 and established therpeutics[5,6,7]
(A) Cumulative percent penetration of nails by HXP124 compared to ciclopirox, tavaborole and efinaconazole.
(B) Cumulative penetration on Day 12 relative to HXP124.

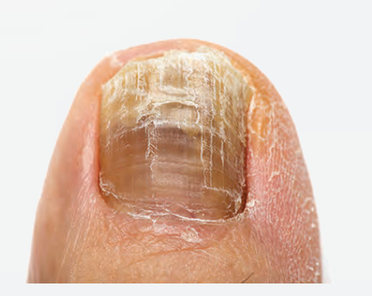

For a copy of this poster please see **hexima.com.au**

[1] Hui *et al.* (2002) *J Pharma Sci* **91**: 189-195
[2] Peraccini *et al.* (2015) *J Fungi* **9**: 30-43
[3] Rosen *et al.* (2015) *J Drugs Dermatol* **14**: 223-228
[4] Grover & Khurana (2012) *Indian J Dermatol Venereol Leprol* **78**: 263
[5] Sugiura *et al.* (2014) *Antimicrob Agents Chemother* **58**: 3837-3842
[6] Hui *et al.* (2007) *J Pharma Sci* **96:** 2622-2631
[7] Gupta *et al.* (2014) *Am J Clin Dermatol* **15**: 489-502
[8] Lay & Anderson (2005) *Curr Protein Pept Sci* **6**: 85-101
[9] Wawrzkiewicz *et al.* (1991) *Mycopathologia* **114**: 1-8
[10] Kyle & Dahl (2004) *Am J Clin Dermatol* **5**: 443-451